3860

Discovery and characterization of novel anti-cancer small molecule inhibitors of Sec61

Eric Lowe[1], Janet L. Anderl[1], Yazmin Carrasco[2], R. Andrea Fan[1], Ying Fang[1], Jing Jiang[1], Henry W.B. Johnson[1], Christopher J. Kirk[1], Dustin McMinn[1], Tony Muchamuel[1], Meera Rao[1], Phillip P. Sharp[2], Jack Taunton[2], Jinhai Wang[1], Jennifer A. Whang[1].

[1]Kezar Life Sciences, Inc. South San Francisco, CA; [2]Department of Cellular and Molecular Pharmacology, University of California, San Francisco, San Francisco, CA

KEZAR LIFE SCIENCES

BACKGROUND

- **The Sec61 translocon is the primary mediator for cotranslational translocation of secreted and transmembrane proteins into the endoplasmic reticulum, providing a novel therapeutic target for blocking expression of therapeutically relevant targets such as cytokines, oncogenic receptors, and immune checkpoint molecules (Figure 1)**
- **Previously described inhibitors of Sec61[2-5] have shown anti-tumor activity but lacked adequate pharmaceutical properties or tolerability for clinical development**
- **Through a drug discovery program to identify novel inhibitors of Sec61 we identified three distinct chemical scaffolds with differential selectivity and potential for treatment of malignant diseases**

Figure 1. Sec61 Translocon[1]

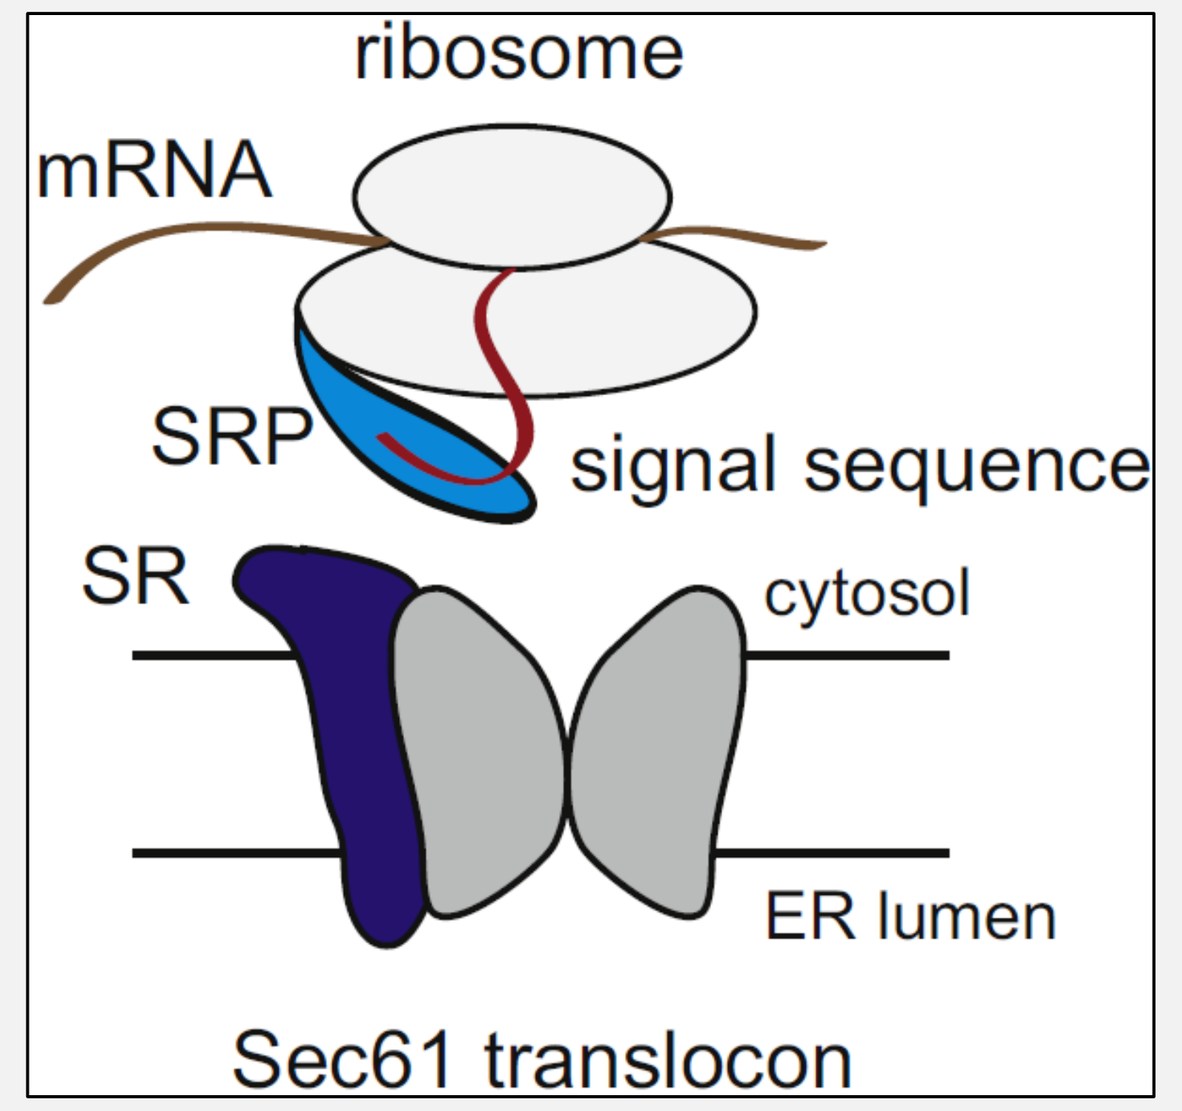
ribosome
mRNA
SRP
signal sequence
SR
cytosol
ER lumen
Sec61 translocon

METHODS

- Inducible luciferase reporter expression plasmids were created by fusing cDNA encoding Gaussia luciferase to the 3' end of signal sequence + 10 amino acids (SS+10AA), transmembrane domain (TMD) or full length (FL) cDNA of proteins of interest. Expression was induced with doxycycline alongside compound treatment for 24 hours (Figures 2-7)
- Mutant R66I[6] or wild type (WT) Sec61 was stably overexpressed in HEK293 cells followed by transient transfection with plasmids expressing luciferase reporters (Figure 3)
- Cell viability was measured by CellTiter-Glo® after the indicated treatment period (CTG; Figures 3, 5, 6, 7)
- HCT116 cells were selected for resistance to CT9 generating R66I mutant Sec61 cells[6]. WT and mutant Sec61 cells were exposed to CT8 (positive control), proteasome inhibitor carfilzomib (negative control), or test compounds for 48 hours followed by viability measurement as described above (Figure 3)
- Luciferase reporter plasmids were transiently transfected into Flp-In T-REx™ 293 cells followed by induction and compound treatment as described above (Figure 4, 8)
- HER3 expression in BT474 breast cancer cells was evaluated after 24 hours of compound treatment by capillary immunoassay (WES-Protein Simple; Figure 6)
- CD4 and PD-1 expression was measured by flow cytometry on T-cells activated with antibodies to CD3 and CD28 alongside compound treatment for 24 hours (Figure 7)
- PD-1, LAG3, TIM3, and CD96 expression was measured by flow cytometry on PBMCs stimulated with antibodies to CD3 and CD28 for 72 hours followed by 24 hour compound treatment (Figure 8)
- Previously frozen human PBMCs and fresh whole blood (n=3 donors) were treated with compound for 20 hours followed by flow cytometry analysis (Figure 9)
- Cytokine levels were measured in human PBMCs (n=3 donors) stimulated with antibodies to CD3 and CD28 (GM-CSF, IFN-γ, TNF-α) or LPS (IL-6, IL-23) alongside compound treatment for 24 hours via Meso Scale Discovery electrochemiluminescent detection (MSD; Figure 9)
- BALB/c mice (n=5) were dosed with KZR-9098 2 hours prior to administration of LPS. Samples were collected 2 hours later and serum TNF-α levels were measured by MSD (Figure 10)
- BALB/c mice were dosed as indicated followed by whole blood collection and flow cytometry analysis 20 hours later (Figure 10)

Screening Workflow

Primary Screen: Inhibitors of Targets 1 & 2 reporters → **Selectivity Screen**: > 40 reporters (e.g. PD-1, IL-2, TNF-α, HER3) → **Counterscreens**: Cytosolic Firefly-luc, Mutant R66I Sec61, Normal cell cytotoxicity → **Potency Evaluation**: Anti-tumor activity, Surface marker expression, Pharmacologic properties

RESULTS

Figure 2. Luciferase Tagged Sec61 Client Proteins Enable Target Selectivity Profiling

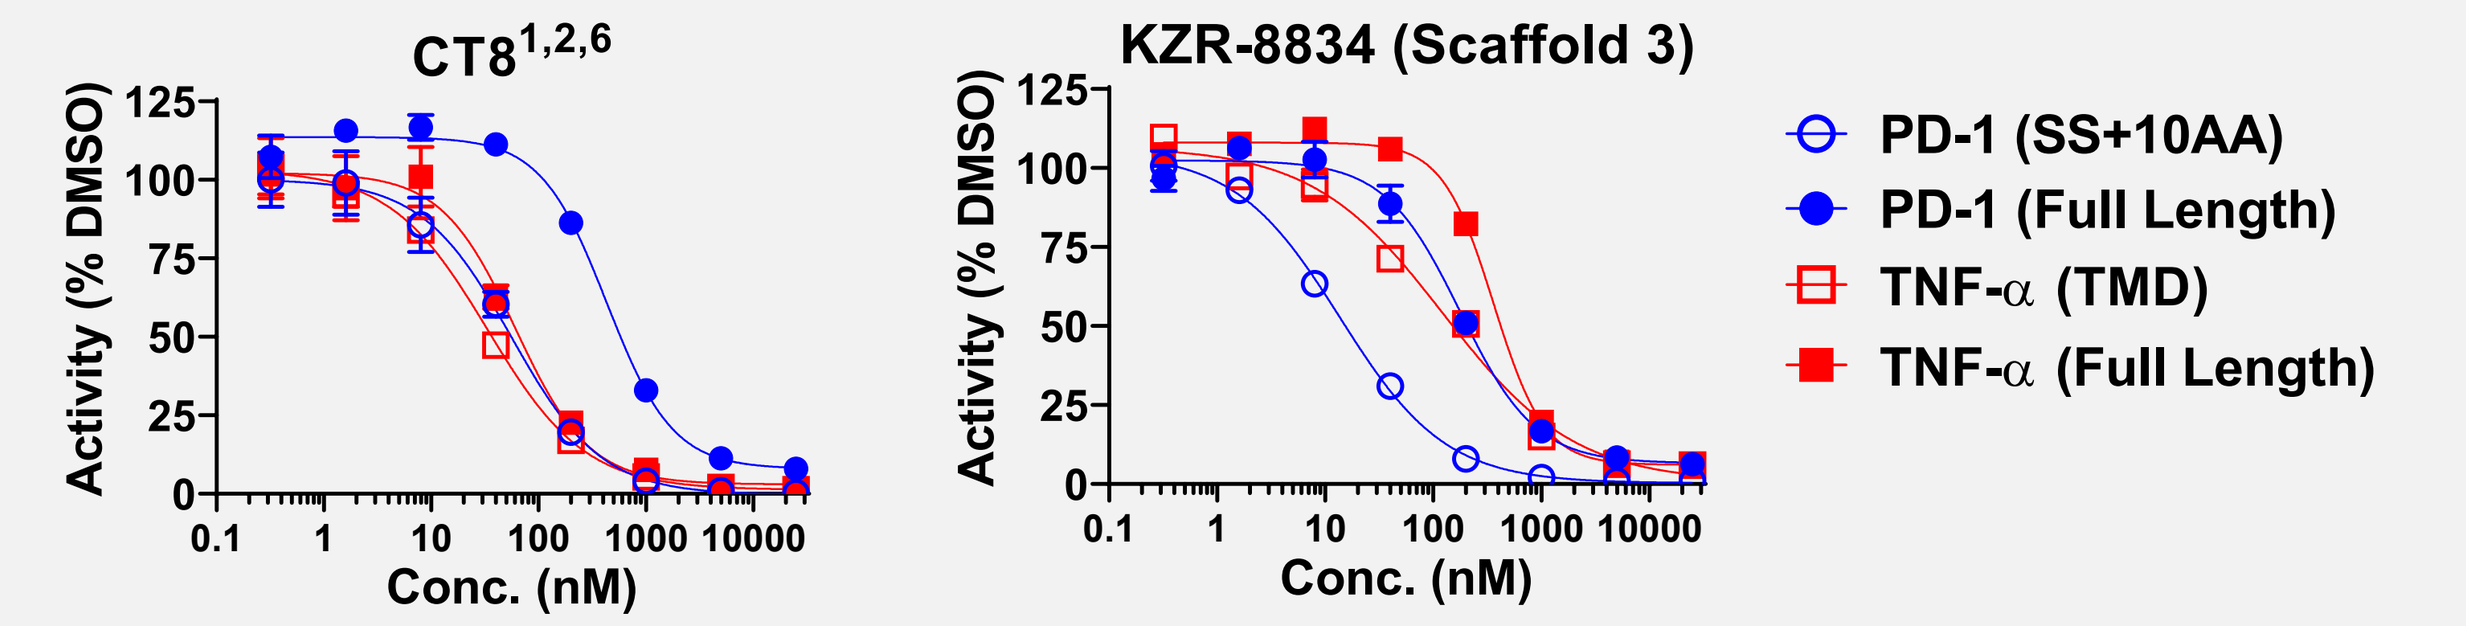
CT8 1,2,6
KZR-8834 (Scaffold 3)
Activity (% DMSO)
Conc. (nM)
PD-1 (SS+10AA)
PD-1 (Full Length)
TNF-α (TMD)
TNF-α (Full Length)

IC_{50} (nM)	CT8	KZR-8834
PD-1 (SS+10AA)	56	14
PD-1 (FL)	431	177
TNF-α (TMD)	33	125
TNF-α (FL)	61	361

Figure 3. Cytosolic Firefly Luciferase and Sec61 Mutant Verify On-Target Inhibition

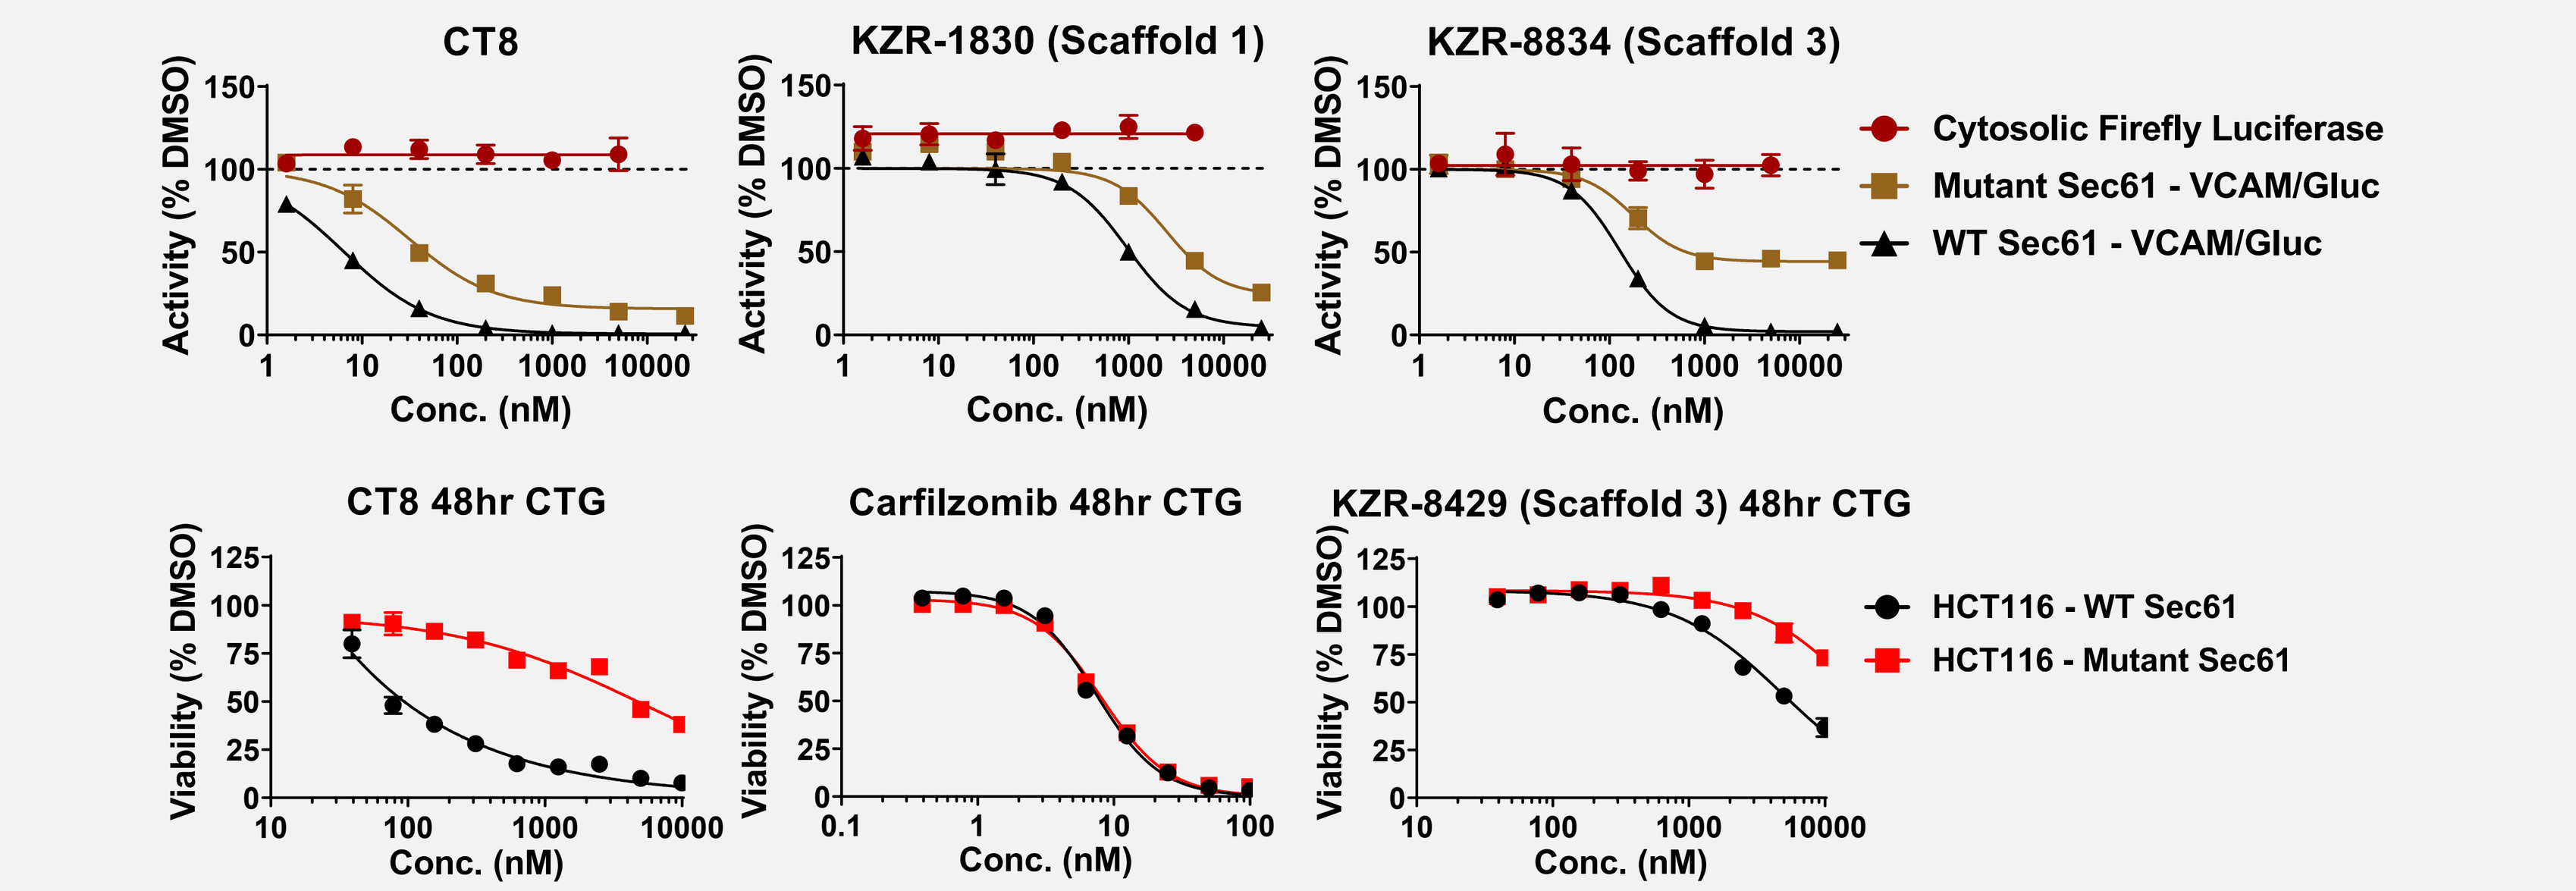
CT8
KZR-1830 (Scaffold 1)
KZR-8834 (Scaffold 3)
Activity (% DMSO)
Conc. (nM)
Cytosolic Firefly Luciferase
Mutant Sec61 - VCAM/Gluc
WT Sec61 - VCAM/Gluc
CT8 48hr CTG
Carfilzomib 48hr CTG
KZR-8429 (Scaffold 3) 48hr CTG
Viability (% DMSO)
HCT116 - WT Sec61
HCT116 - Mutant Sec61

Figure 4. Distinct Chemical Scaffolds Achieve Differential Sec61 Client Selectivity

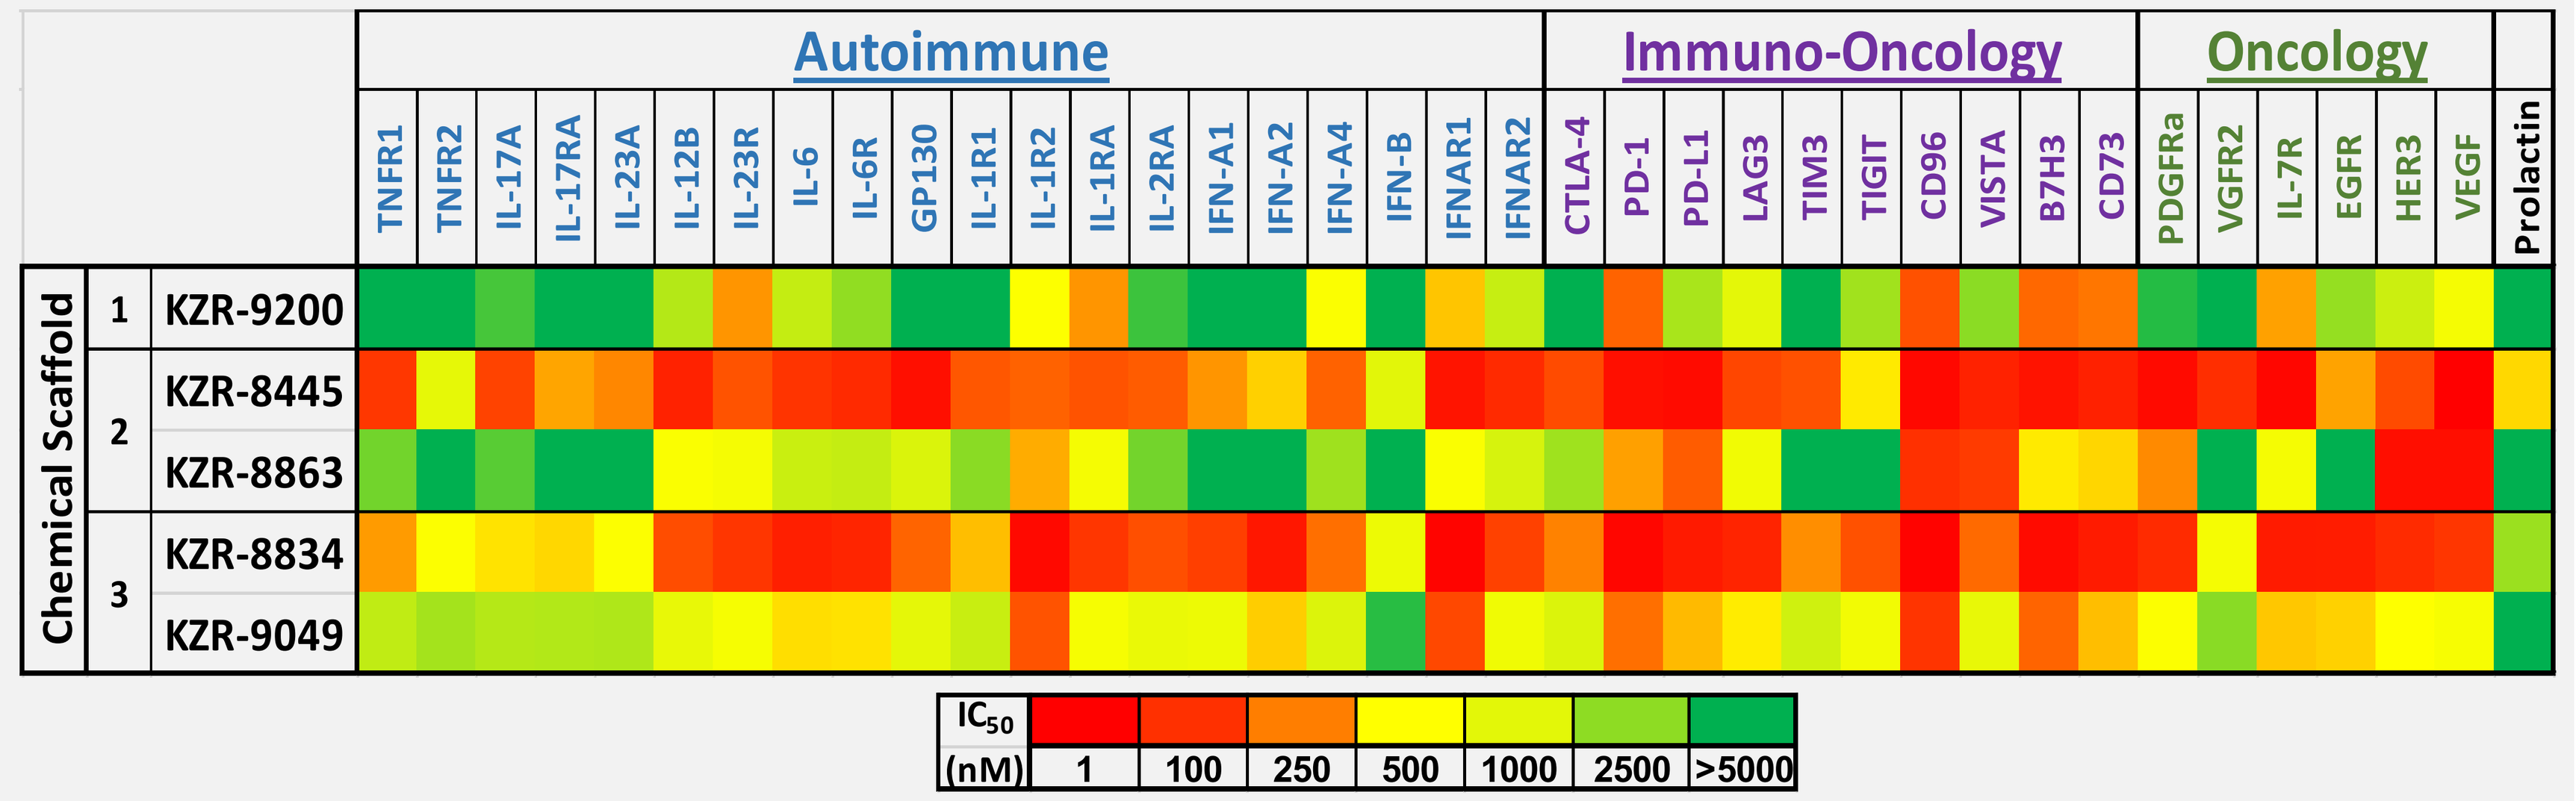
Autoimmune
Immuno-Oncology
Oncology
TNFR1 TNFR2 IL-17A IL-17RA IL-23A IL-12B IL-23R IL-6 IL-6R GP130 IL-1R1 IL-1R2 IL-1RA IL-2RA IFN-A1 IFN-A2 IFN-A4 IFN-B IFNAR1 IFNAR2
CTLA-4 PD-1 PD-L1 LAG3 TIM3 TIGIT CD96 VISTA B7H3 CD73
PDGFRa VGFR2 IL-7R EGFR HER3 VEGF
Prolactin
Chemical Scaffold
1 KZR-9200
2 KZR-8445
KZR-8863
3 KZR-8834
KZR-9049
IC50 (nM) 1 100 250 500 1000 2500 >5000

Figure 5. Scaffold 1: Broad Sec61 Inhibition and Anti-Cancer Activity or Target (VCAM1) Selective Inhibition

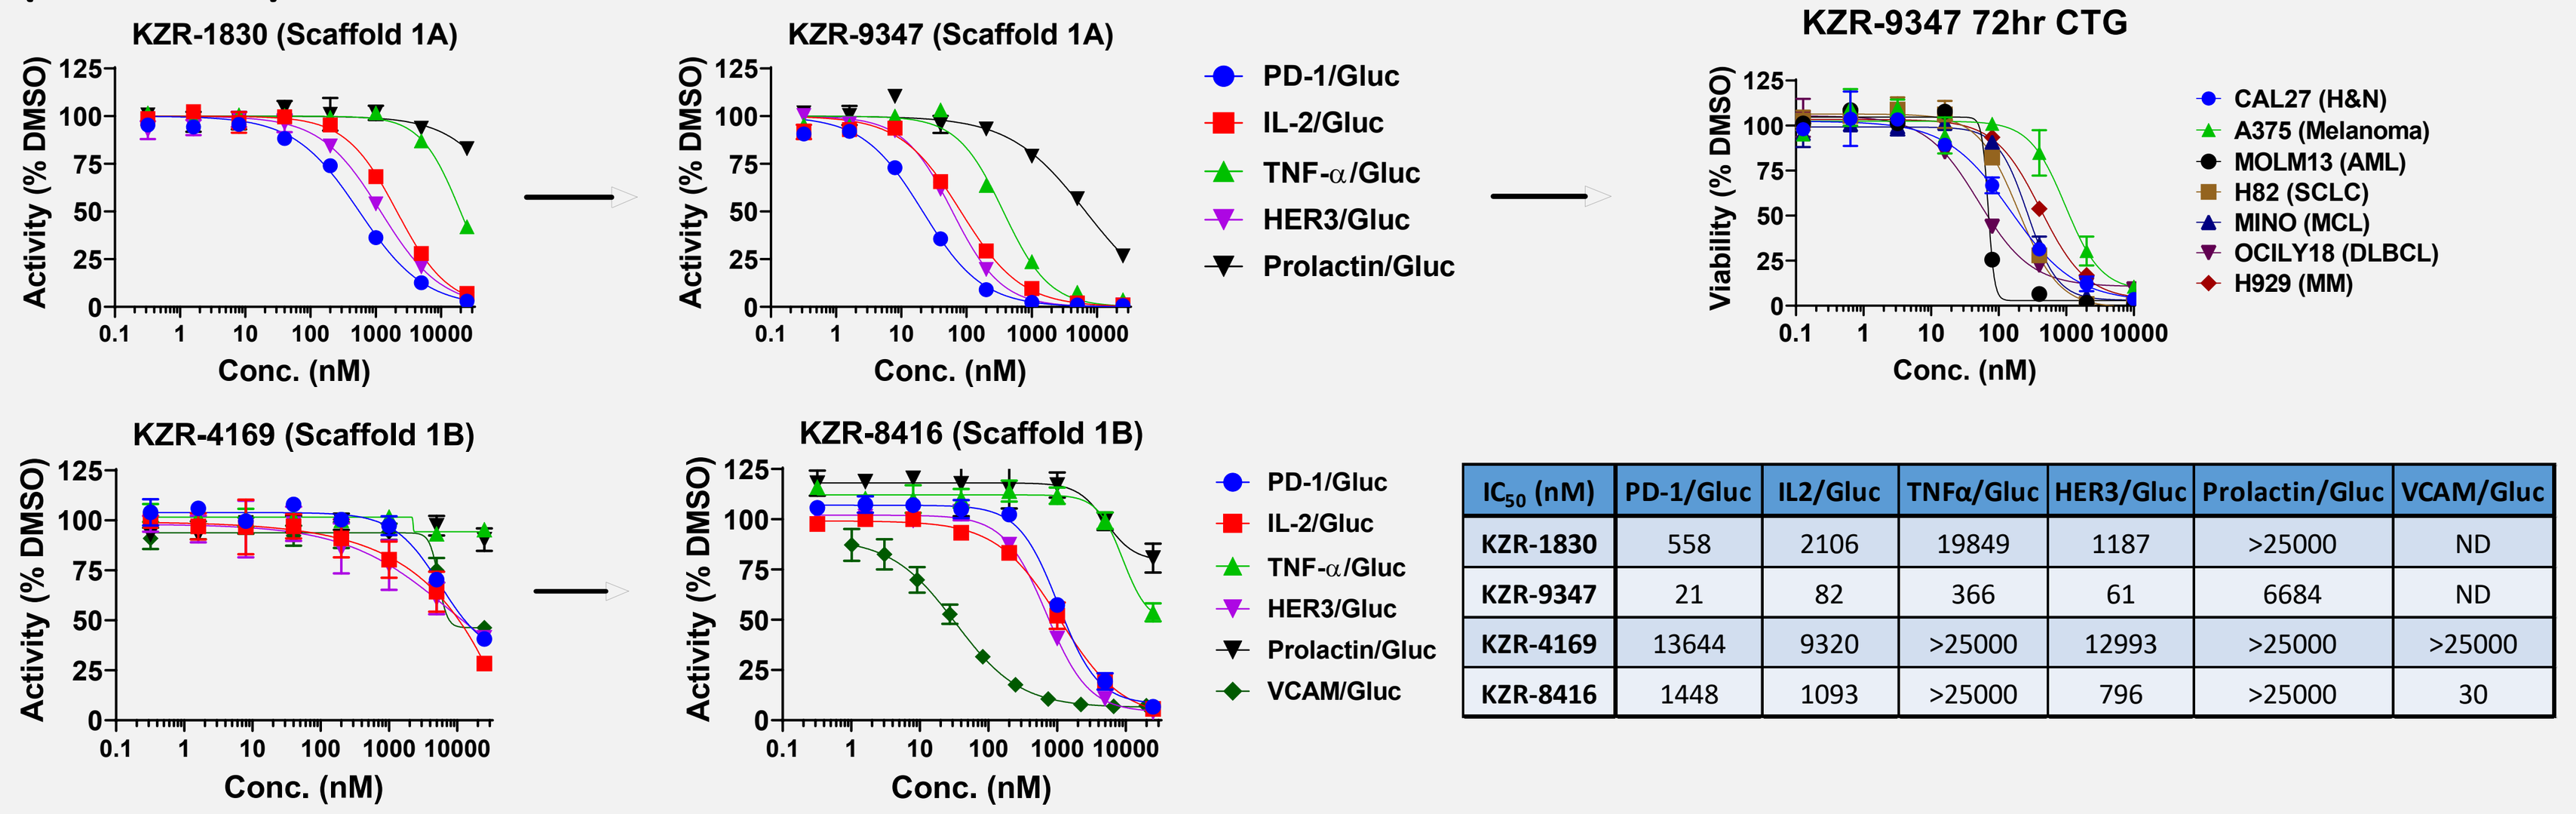
KZR-1830 (Scaffold 1A)
KZR-9347 (Scaffold 1A)
KZR-9347 72hr CTG
KZR-4169 (Scaffold 1B)
KZR-8416 (Scaffold 1B)
Activity (% DMSO)
Viability (% DMSO)
Conc. (nM)
PD-1/Gluc
IL-2/Gluc
TNF-α/Gluc
HER3/Gluc
Prolactin/Gluc
VCAM/Gluc
CAL27 (H&N)
A375 (Melanoma)
MOLM13 (AML)
H82 (SCLC)
MINO (MCL)
OCILY18 (DLBCL)
H929 (MM)

IC_{50} (nM)	PD-1/Gluc	IL2/Gluc	TNFα/Gluc	HER3/Gluc	Prolactin/Gluc	VCAM/Gluc
KZR-1830	558	2106	19849	1187	>25000	ND
KZR-9347	21	82	366	61	6684	ND
KZR-4169	13644	9320	>25000	12993	>25000	>25000
KZR-8416	1448	1093	>25000	796	>25000	30

Figure 6. Scaffold 2: Broad Anti-Cancer Activity or Selective HER3 Blockade

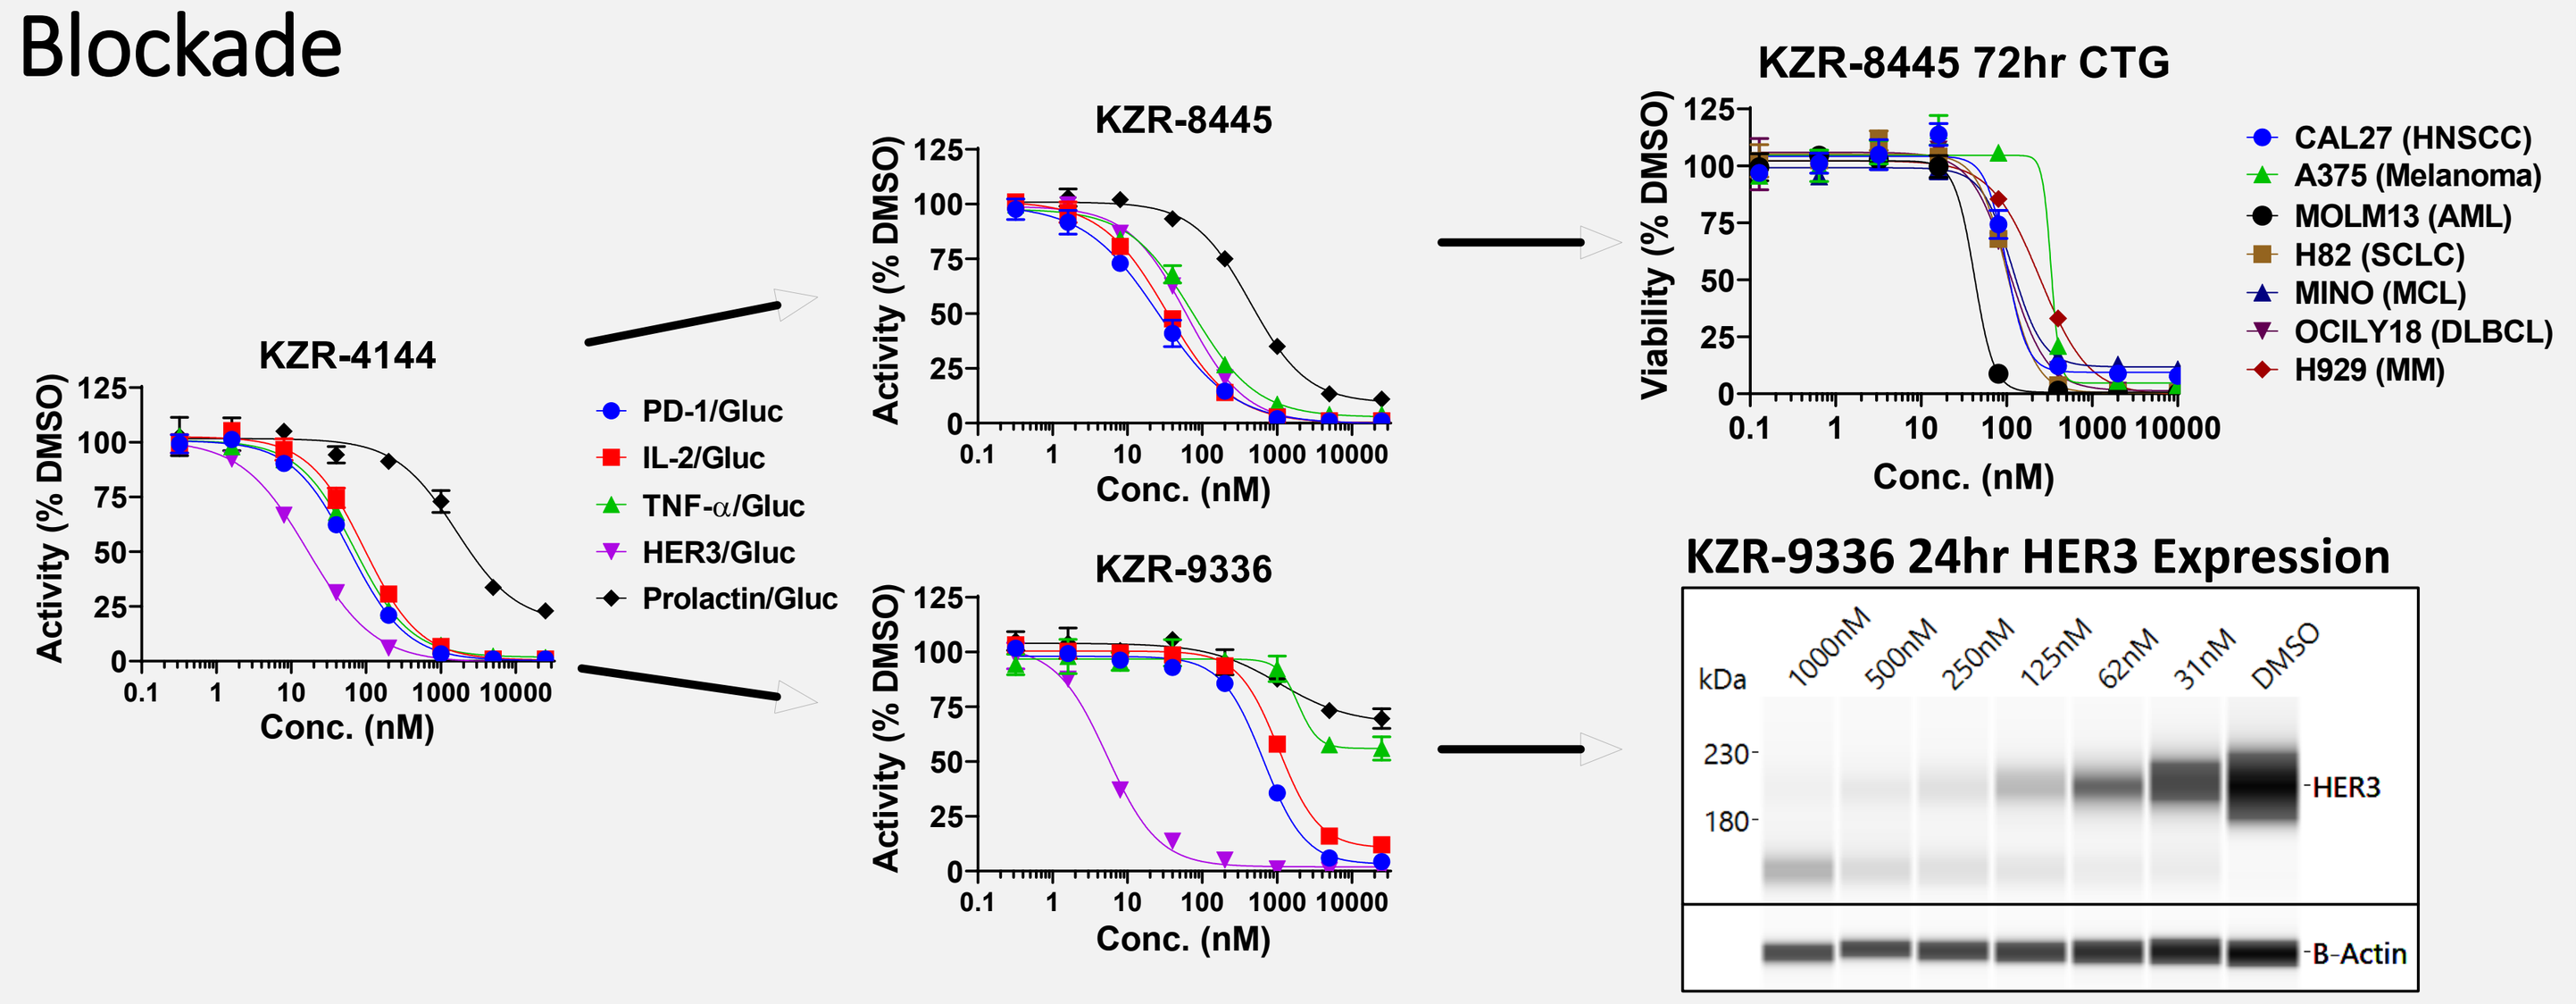
KZR-4144
KZR-8445
KZR-9336
KZR-8445 72hr CTG
KZR-9336 24hr HER3 Expression
Activity (% DMSO)
Viability (% DMSO)
Conc. (nM)
PD-1/Gluc
IL-2/Gluc
TNF-α/Gluc
HER3/Gluc
Prolactin/Gluc
CAL27 (HNSCC)
A375 (Melanoma)
MOLM13 (AML)
H82 (SCLC)
MINO (MCL)
OCILY18 (DLBCL)
H929 (MM)
kDa
1000nM 500nM 250nM 125nM 62nM 31nM DMSO
230
180
HER3
B-Actin

Figure 7. Scaffold 3: Broad Anti-Cancer Activity or Selective PD-1 Inhibition

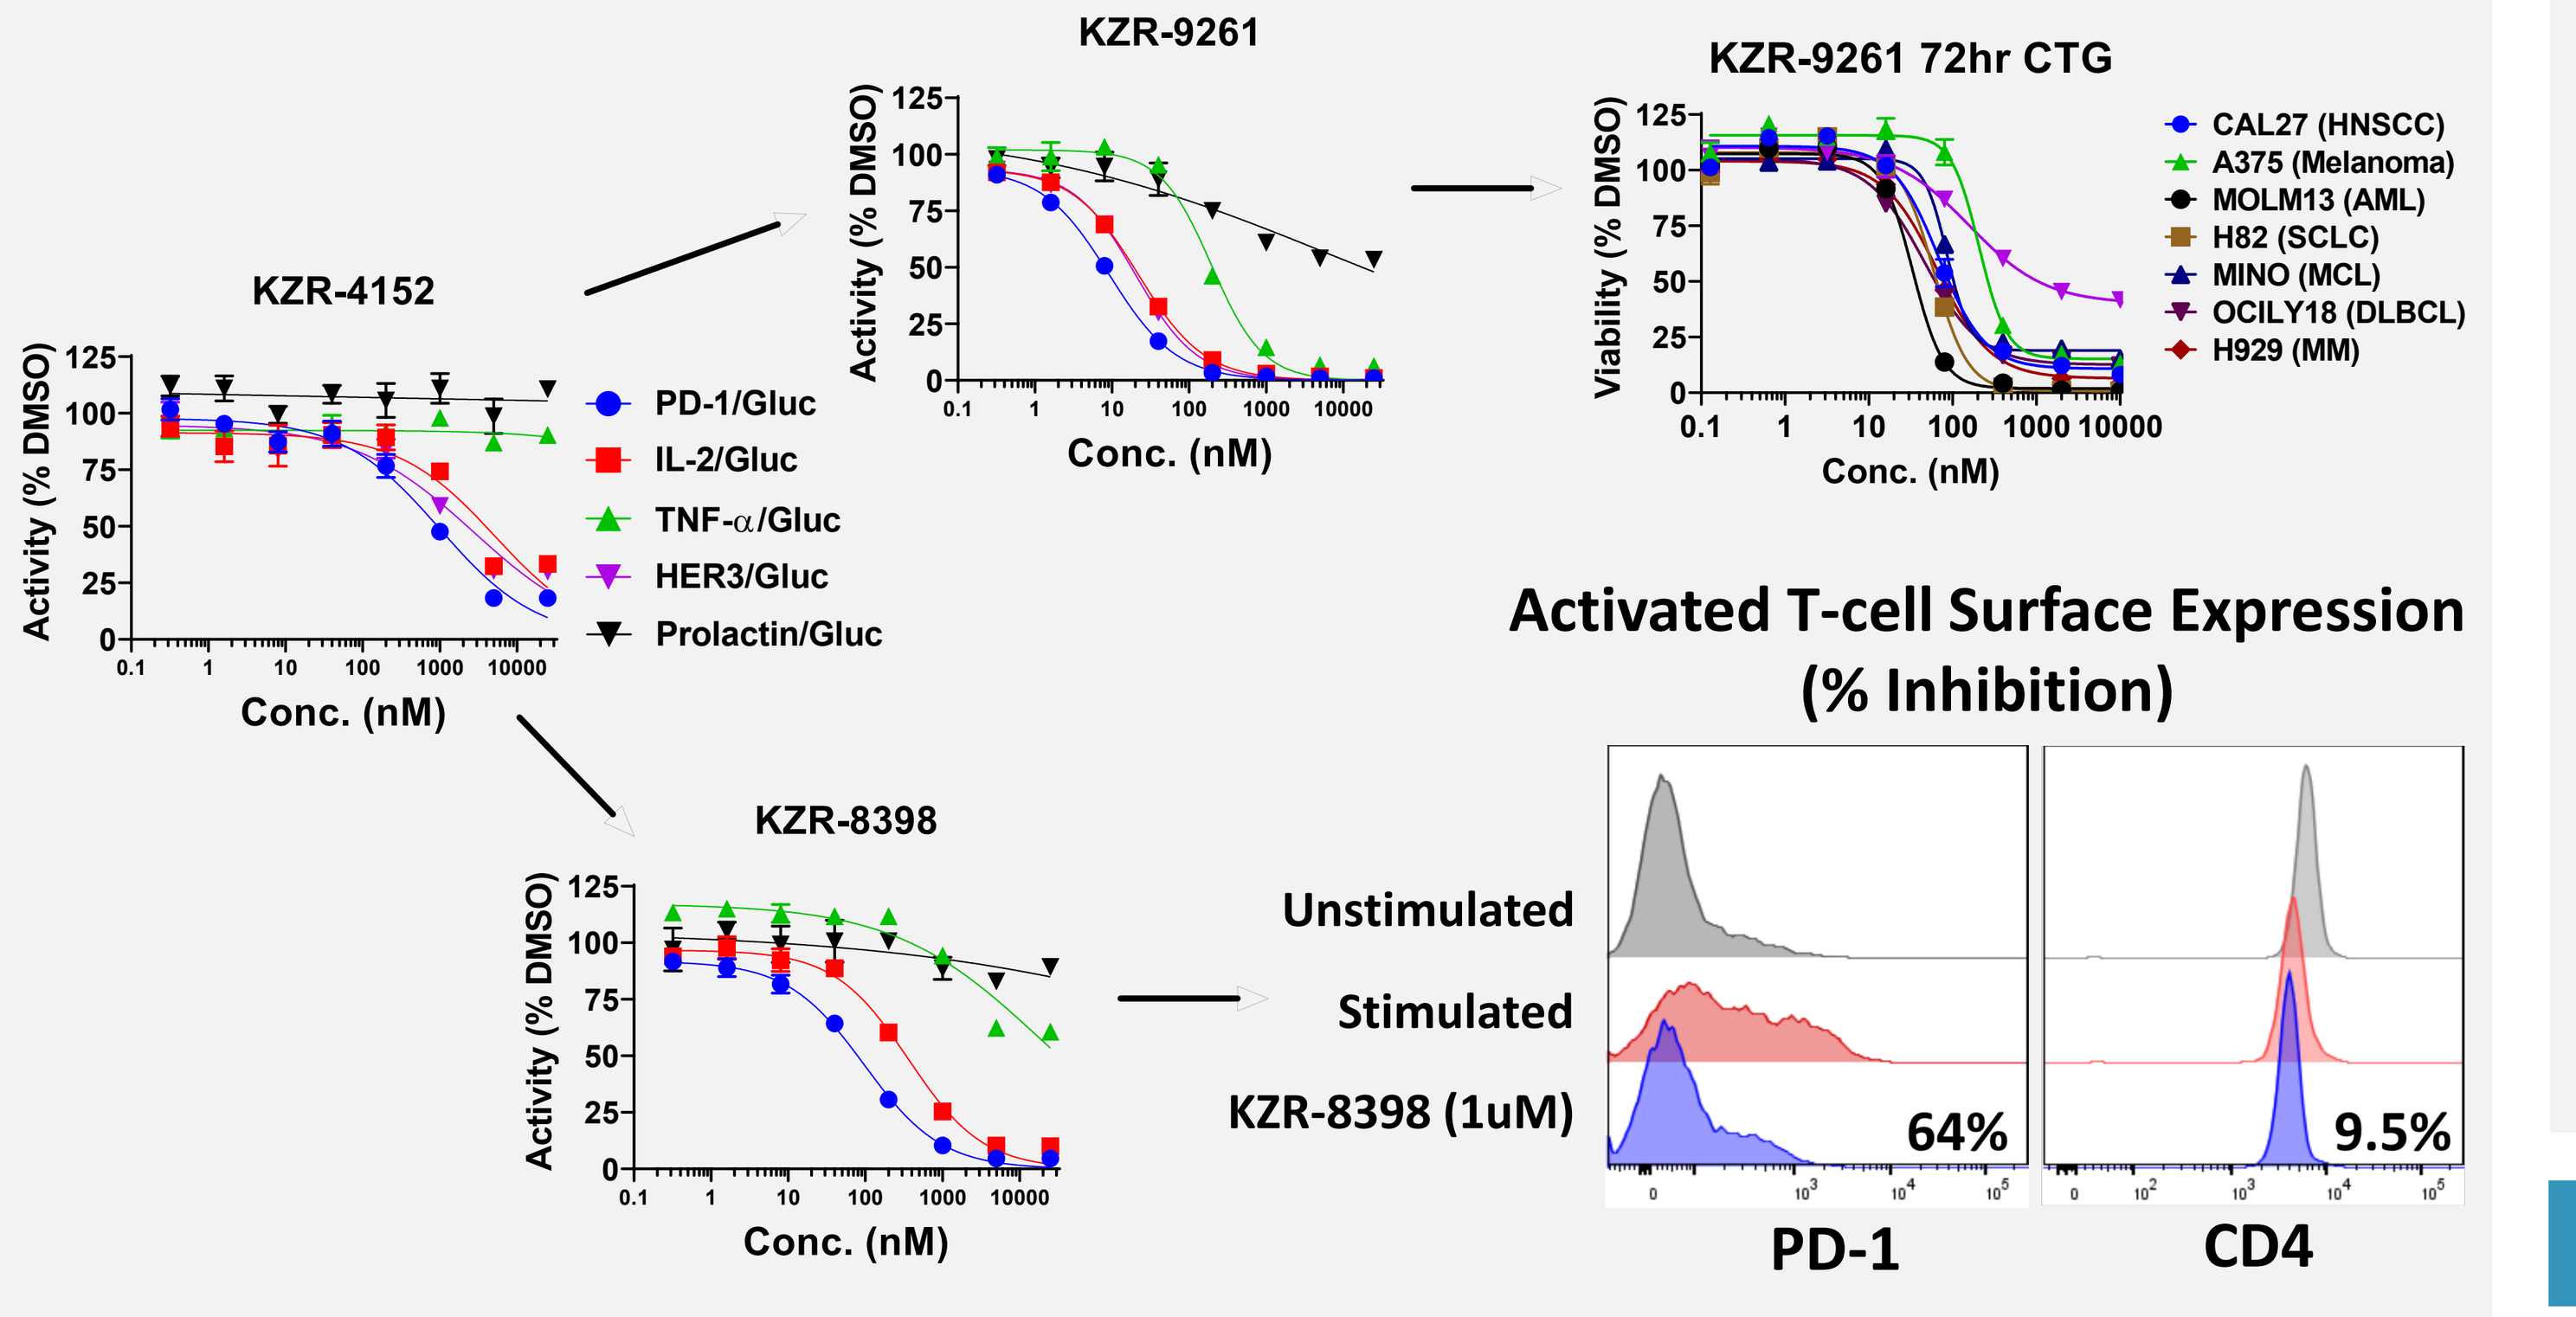
KZR-4152
KZR-9261
KZR-9261 72hr CTG
KZR-8398
Activity (% DMSO)
Viability (% DMSO)
Conc. (nM)
PD-1/Gluc
IL-2/Gluc
TNF-α/Gluc
HER3/Gluc
Prolactin/Gluc
CAL27 (HNSCC)
A375 (Melanoma)
MOLM13 (AML)
H82 (SCLC)
MINO (MCL)
OCILY18 (DLBCL)
H929 (MM)
Activated T-cell Surface Expression (% Inhibition)
Unstimulated
Stimulated
KZR-8398 (1uM)
64%
9.5%
PD-1
CD4

Figure 8. Scaffolds 1 and 3 Exhibit Potent Blockade of Multiple Immune Checkpoint Proteins

Immune Checkpoint Luciferase Reporter IC_{50} (nM)

Chemical Scaffold			CTLA-4	PD-1	PD-L1	LAG3	TIM3	TIGIT	CD96	VISTA	B7H3	CD73	CD47
	1	KZR-9275	1261	106	500	260	2331	933	62	345	68	25	16
	3	KZR-9039	8	72	20	41	90	90	7	138	10	29	32

PBMC Surface Expression (% Inhibition)

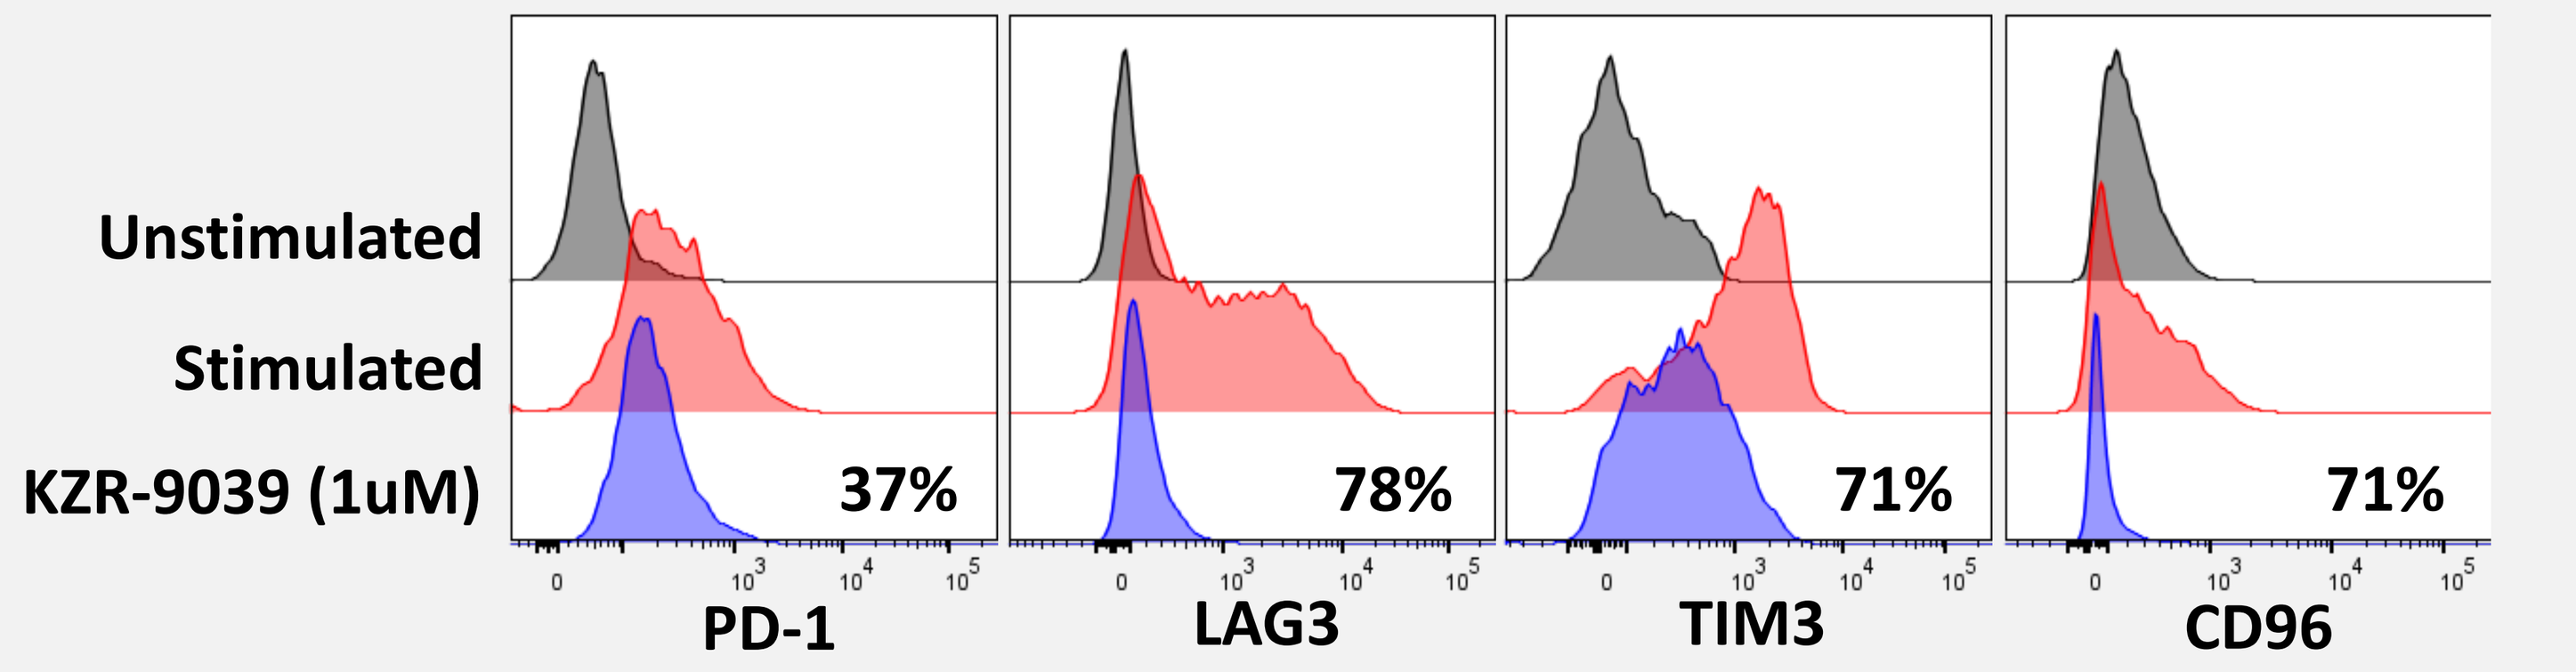
Unstimulated
Stimulated
KZR-9039 (1uM)
37%
78%
71%
71%
PD-1
LAG3
TIM3
CD96

Figure 9. Inhibition of Sec61 Clients in Primary Cell Assays

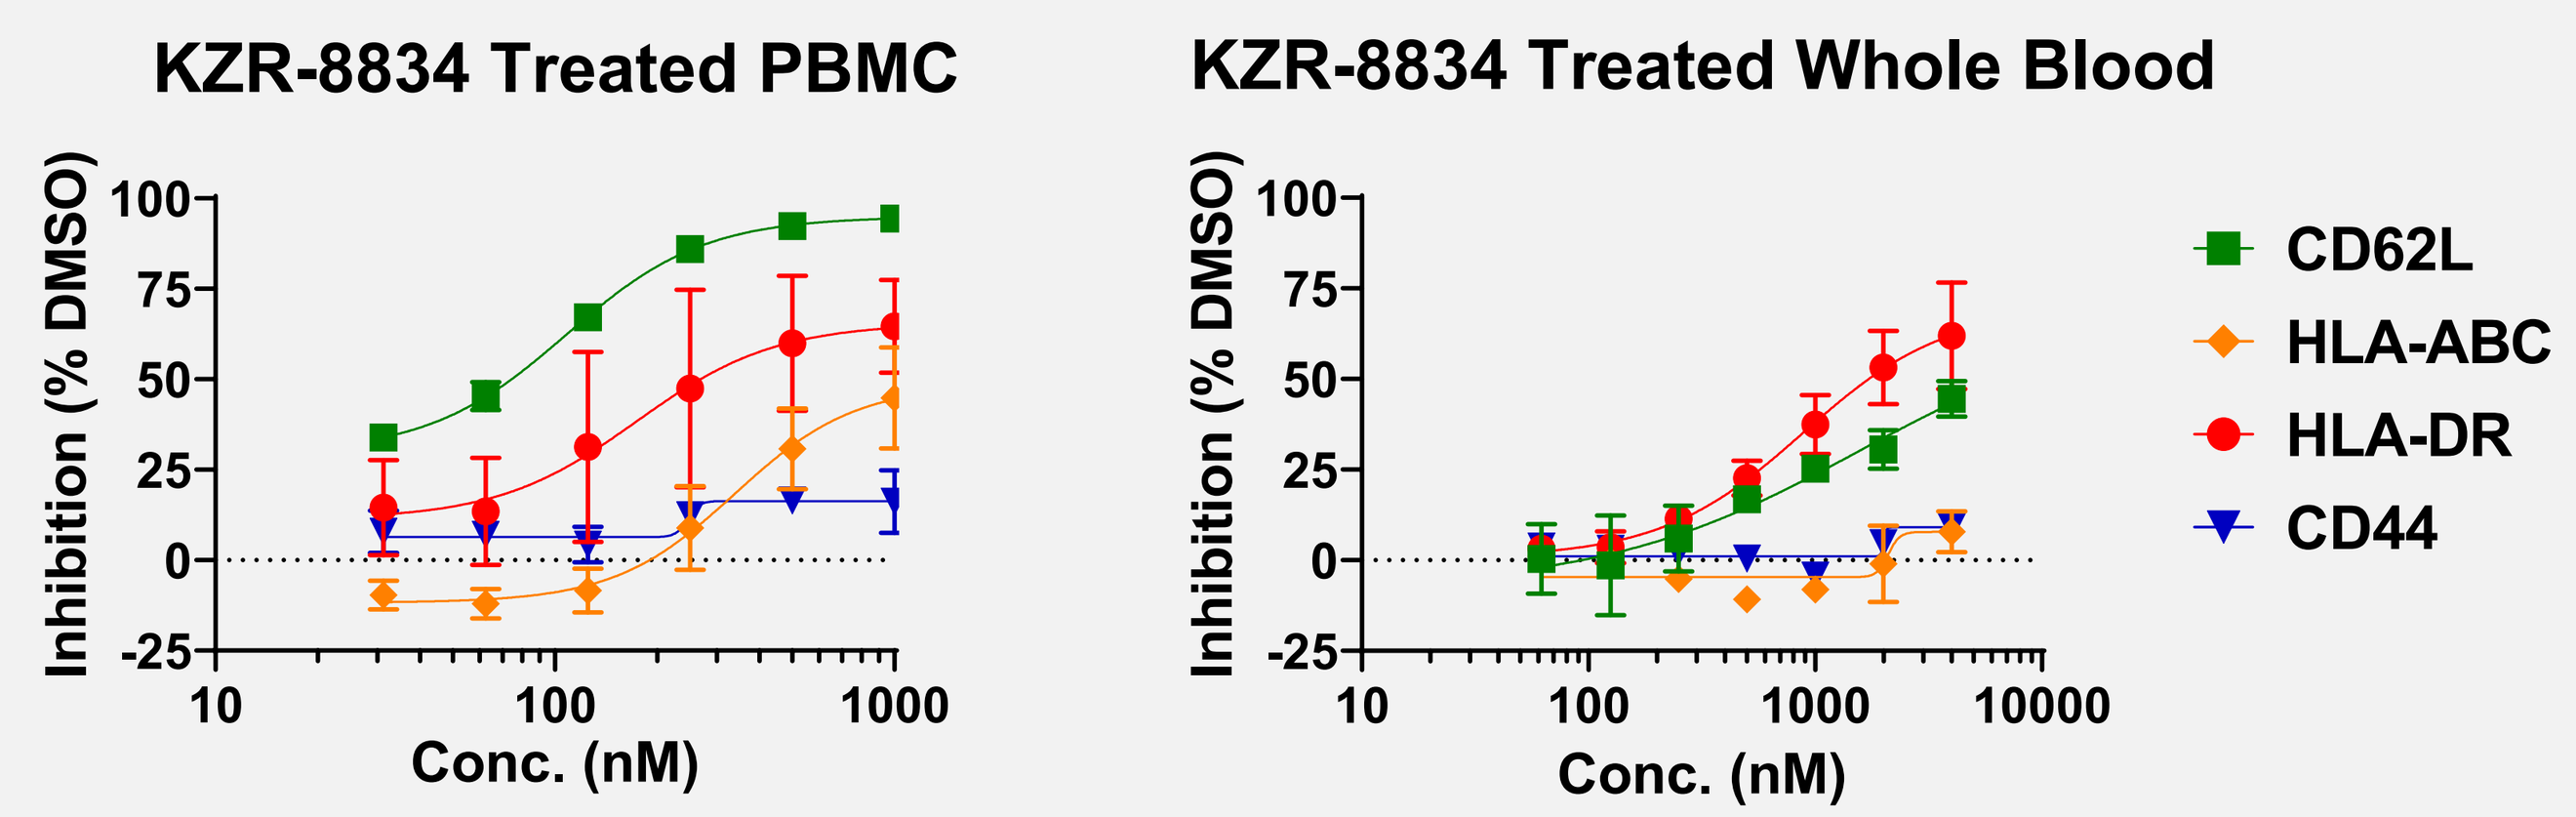
KZR-8834 Treated PBMC
KZR-8834 Treated Whole Blood
Inhibition (% DMSO)
Conc. (nM)
CD62L
HLA-ABC
HLA-DR
CD44

Stimulated PBMC Cytokine Levels (Mean IC_{50} (nM))

GM-CSF	IFNγ	TNFα	IL-6	IL-23
165	152	82	61	200

Figure 10. Modulation of Basal and Inducible Sec61 Client Proteins in Mice

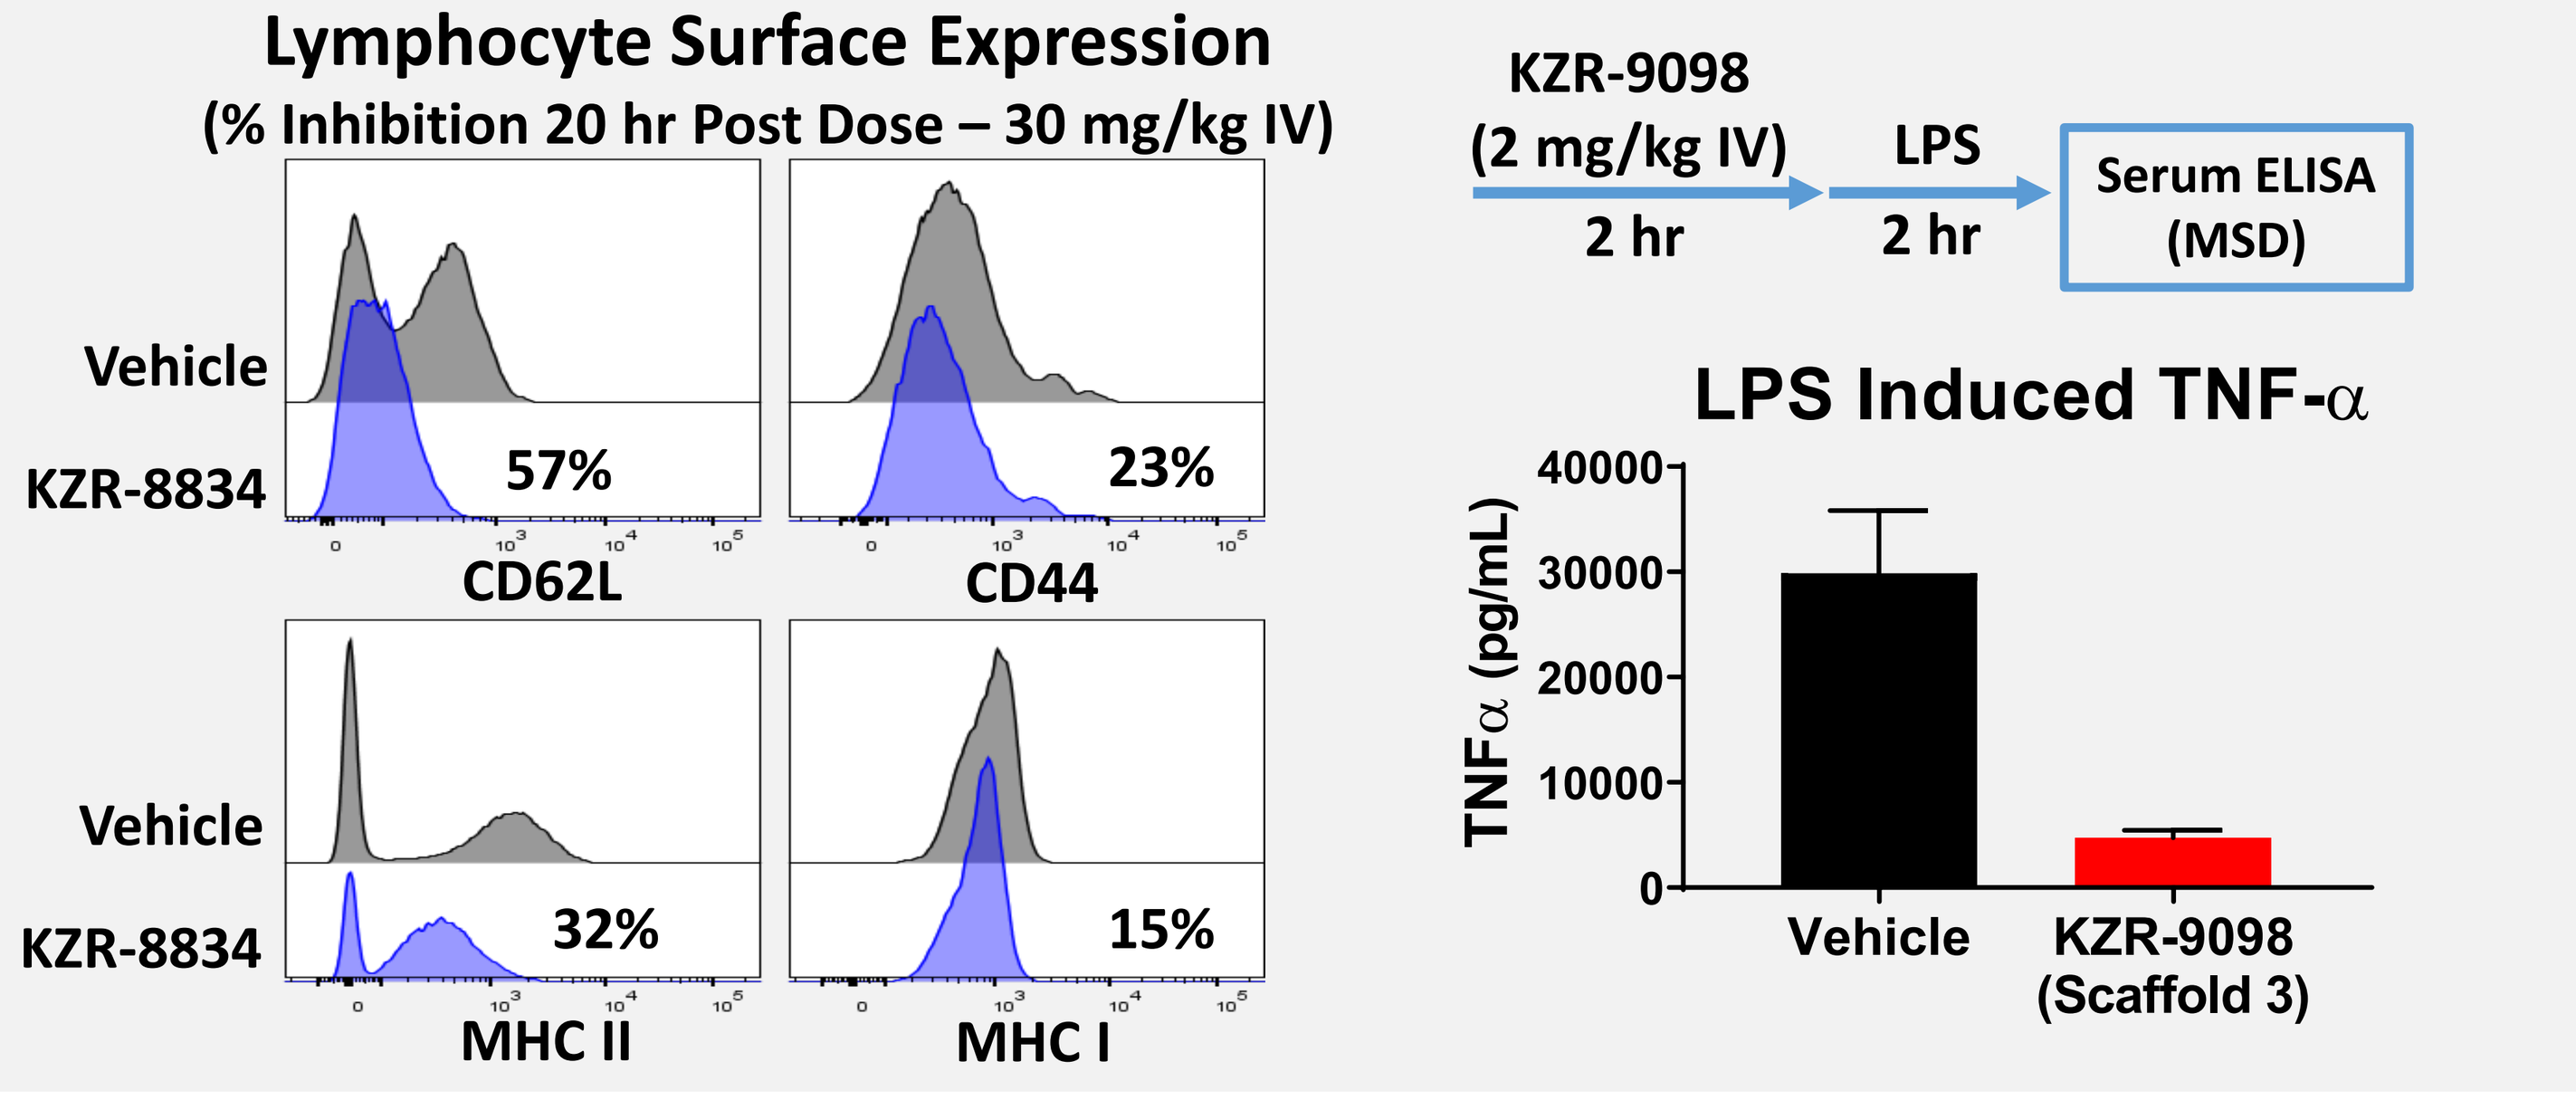
Lymphocyte Surface Expression
(% Inhibition 20 hr Post Dose – 30 mg/kg IV)
Vehicle
KZR-8834
57%
23%
CD62L
CD44
32%
15%
MHC II
MHC I
KZR-9098
(2 mg/kg IV)
2 hr
LPS
2 hr
Serum ELISA
(MSD)
LPS Induced TNF-α
TNFα (pg/mL)
Vehicle
KZR-9098
(Scaffold 3)

CONCLUSIONS

- **Reporter-based screening assays were successfully developed to discover broad and selective inhibitors of Sec61 client proteins**
- **Small molecule targeting of therapeutically relevant cell surface and secreted proteins can be achieved in-vitro and in-vivo**
- **Multiple chemical scaffolds are able to act as broad and selective inhibitors of Sec61 function resulting in reduced expression of key targets in autoimmune/inflammatory, immuno-oncology, and oncology indications**

REFERENCES

1. Maifeld, et al. *Cell Chem. & Bio.* 2011, 18: 1082
2. Garrison, et al. *Nature.* 2005, 436: 285
3. Paatero, et al. *Cell Chem. & Bio. 2016, 23: 561*
4. Juune, et al. *J. Cell Sci.* 2015, 128: 1217
5. Baron, et al. *J. Exp. Med.* 2016, 213: 2885
6. MacKinnon, et al. *eLIFE.* 2014, 3:e01483