Poor Health-Related Quality Of Life In Patients With Lupus Nephritis

Cristina Arriens,[1] Janice Ma,[2] Konrad Pisarczyk,[2] Richard Leff,[3] Kiruthi Palaniswamy,[3] Li Long[3]

[1]Oklahoma Medical Research Foundation & University of Oklahoma Health Science Center, Oklahoma City, Oklahoma, USA [2]Maple Health Group LLC, New York, New York, USA [3]Kezar Life Sciences, Inc. South San Francisco, California, USA

Background/Objectives

- LN is a serious manifestation of SLE, and the disease and its treatments come with considerable morbidity and impact on patients.
- Disease outcomes tend to be worse for patients with refractory LN,[1,2] but little is known about true impact of LN on health-related quality of life (HRQoL) in patients with SLE.
- This study reviews and qualitatively summarizes evidence on the humanistic burden of LN in adult and juvenile patients.

Methods

- A targeted literature review (TLR) was conducted in MEDLINE/Pubmed and Embase in adult and juvenile patients with diagnosed LN.
- Search strategies were developed to identify relevant peer-reviewed articles published in English between March 2012 and March 2022, and conference abstracts indexed in Embase since 2019.
- All records were screened by a single reviewer according to pre-specified inclusion and exclusion criteria.
- The current poster summarizes data on humanistic burden of disease and treatment impact on HRQoL in adults and children with LN.

Results

- Of the 4,216 records identified in the medical databases, 11 non-interventional studies evaluated impact of LN on patients' HRQoL.[3-8,11-15]
- In addition, 1 randomized controlled trial (RCT) and 1 real-world evidence (RWE) study assessed HRQoL outcomes associated with the current standard of care (SoC) therapy in LN patients.[9,10]
- Twelve studies included adults and one study was focused on juvenile patients with LN **(Figure 1).**

Figure 1. PRISMA diagram

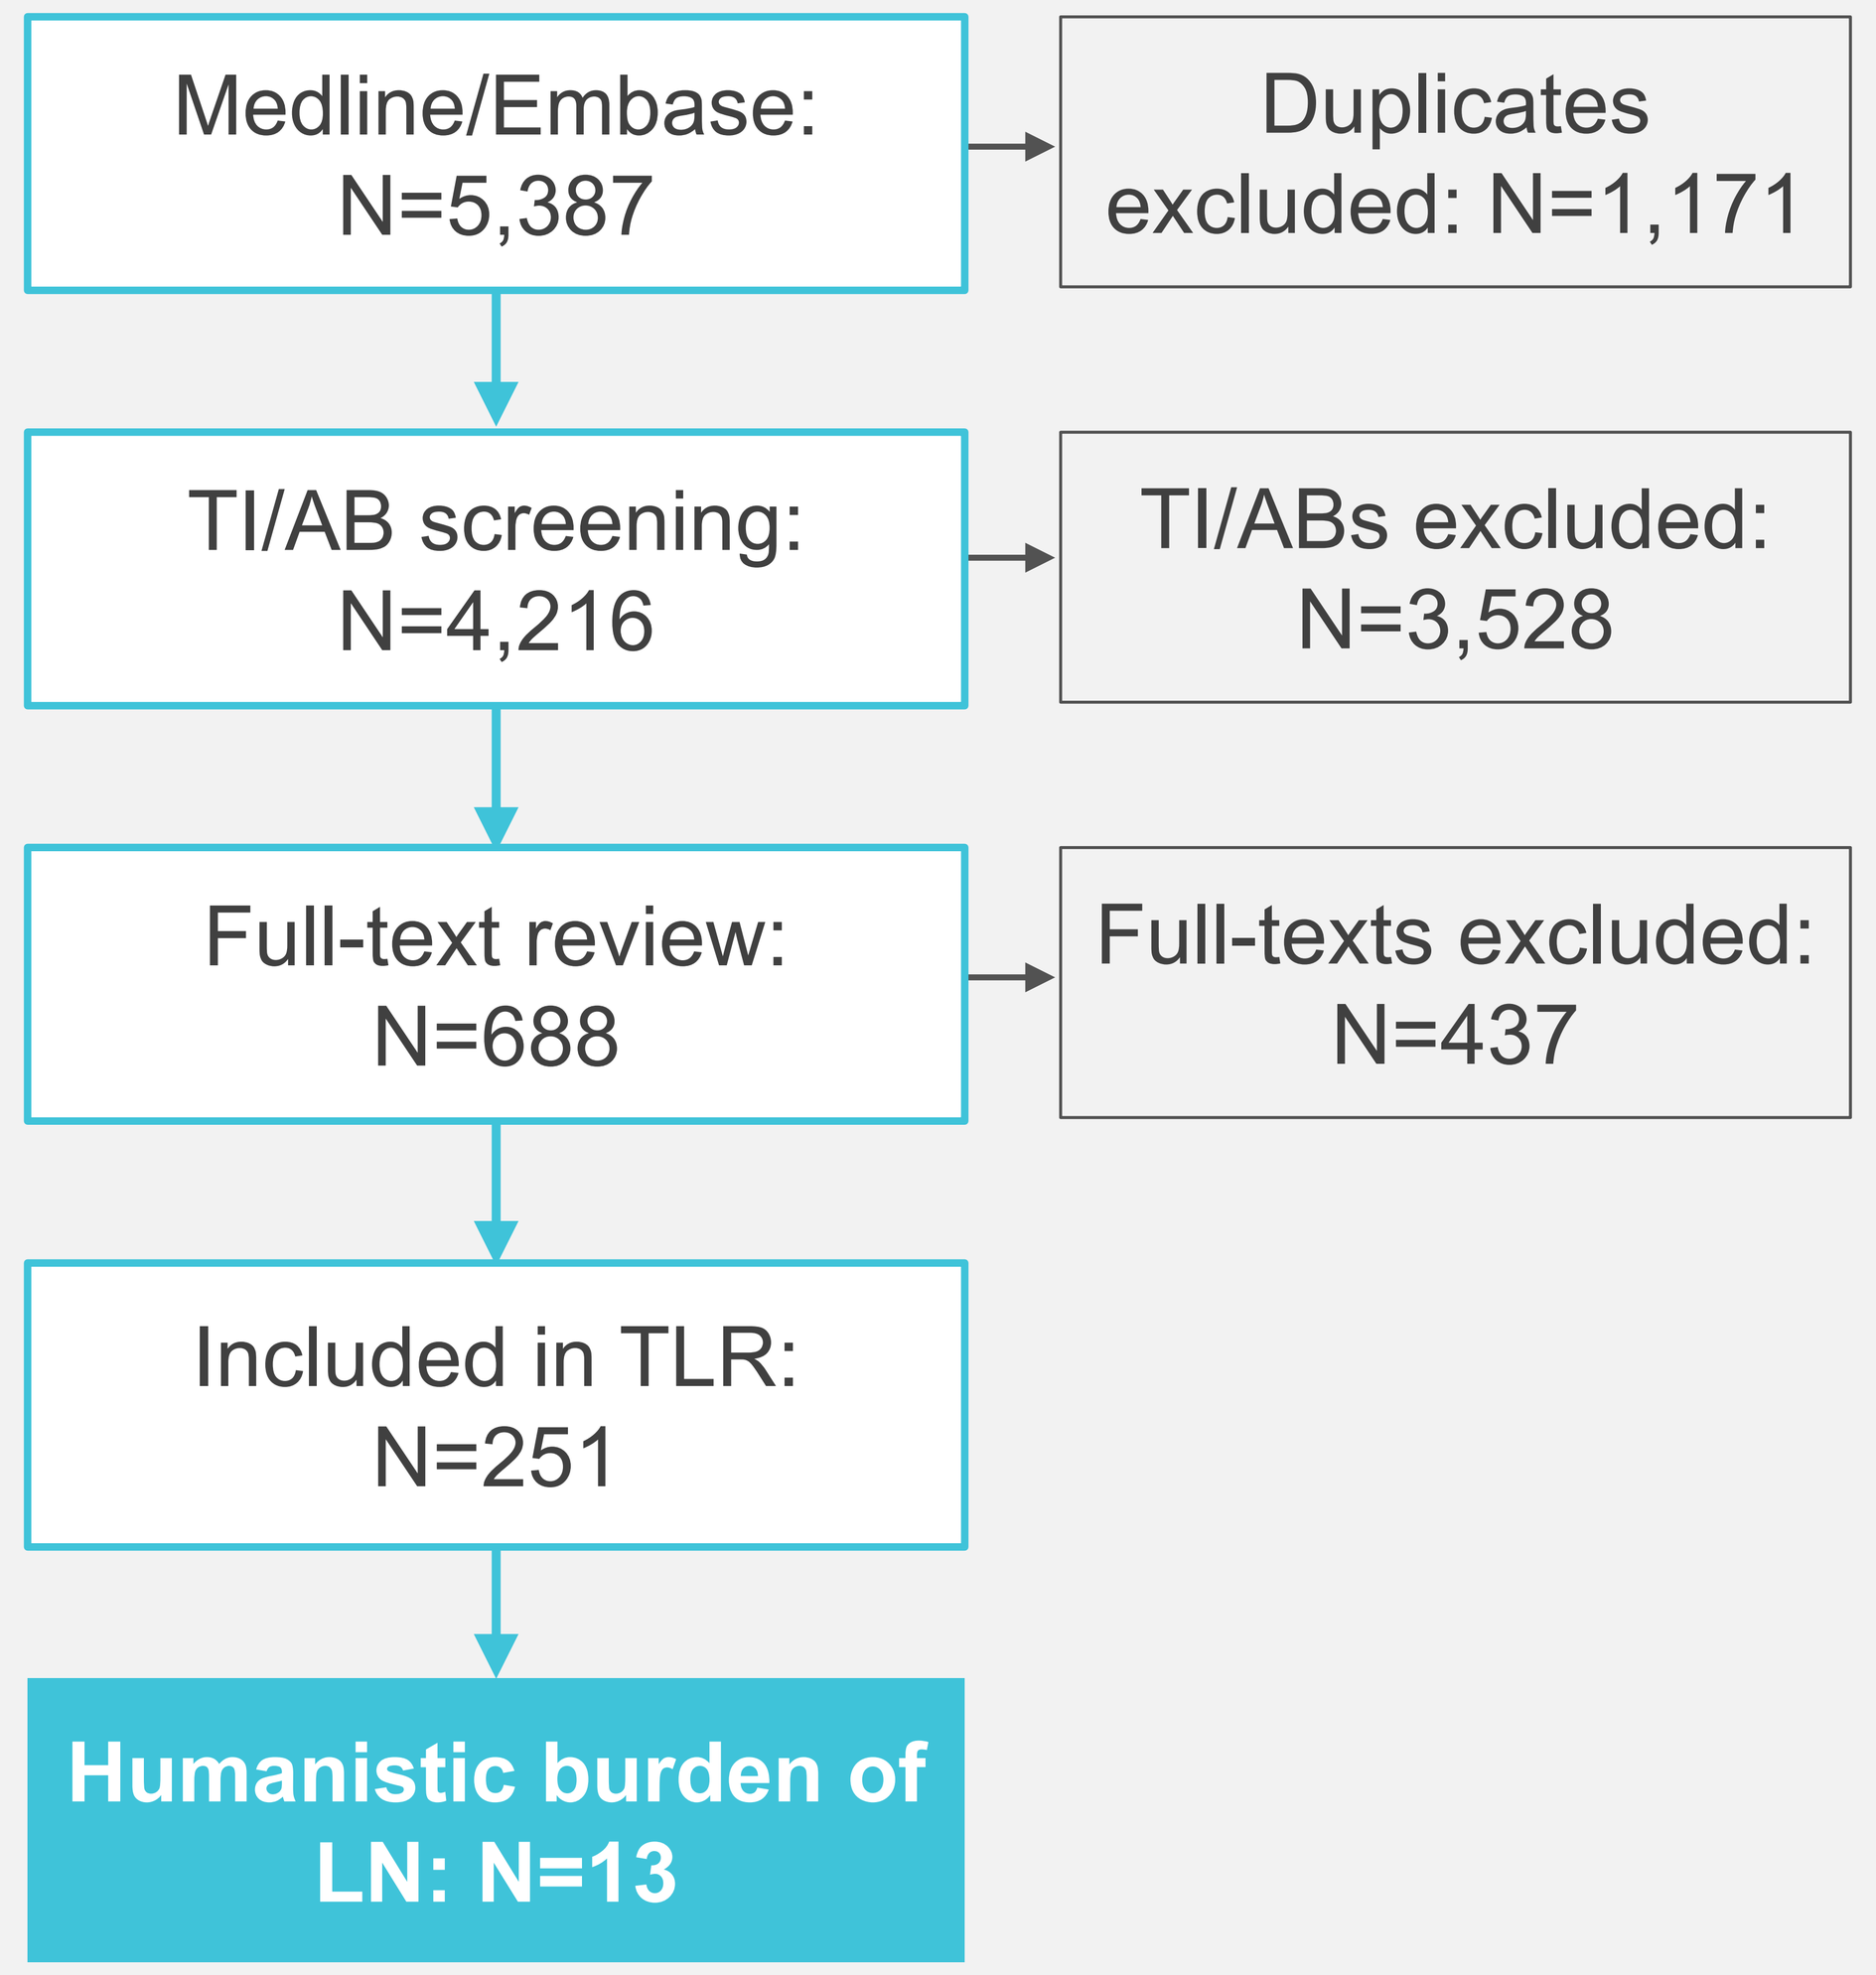
Medline/Embase: N=5,387
Duplicates excluded: N=1,171
TI/AB screening: N=4,216
TI/ABs excluded: N=3,528
Full-text review: N=688
Full-texts excluded: N=437
Included in TLR: N=251
Humanistic burden of LN: N=13

- Six studies included patients from the USA. The remaining studies were conducted in multiple countries worldwide (n=3), Latin America (n=1), the Netherlands (n=1), Switzerland (n=1), and Romania (n=1).

Results (cont'd)

Burden of Active LN

- Active LN had the greatest HRQoL impact on the domains of procreation, physical, and emotional functioning[3-6] compared to patients with inactive LN or SLE-only, as measured by lupus-specific questionnaires (LupusPRO, LupusQoL, etc.) **(Table 1)**.
- Patient-perceived burden of active LN was more pronounced vs. other SLE organ system manifestations, with adults with active LN reporting poor scores in almost all domains of the SF-36,[7,8] suggesting a deterioration in multiple aspects of life, such as physical and emotional functioning **(Table 1)**.

Table 1. Adults with LN, especially those with active LN, had significantly worse HRQoL in various lupus-specific questionnaires and SF-36 domains compared to patients with SLE-only or those with inactive LN

	Study Cohort	Control Cohort	LN vs Control
	LUPUS SPECIFIC QUESTIONNAIRES		
GLOBAL	**SLE + LN (N=539)[3]**	SLE only (N=718)	**Worse scores** in LupusPRO medications, procreation, pain-vitality domains ($p<0.03$)
USA	**SLE + LN (N=34)[5]**	SLE only (N=138)	Lower prevalence of moderate-to-severe forgetfulness, muscle pain, and numbness in SLAQ assessment ($p<0.05$)
GLOBAL	**SLE + Active LN (N=129)[3]**	SLE + Inactive LN (N=410)	**Worse scores** in LupusPRO symptoms, medications, procreation, emotional health, body image, and summary HRQoL domains ($p<0.01$)
LATAM	**SLE + Active LN (N=294)[4]**	SLE + Inactive LN (N=204)	**Worse scores** in all domains of LupusQoL ($p\leq0.001$)
USA	**SLE + Active LN (N=34)[5]**	SLE + Inactive LN (N=33)	**Worse SLAQ scores** in disease severity and muscle pain ($p<0.05$)
USA	**SLE and/or LN with ≥1 flare/year (N=904)[6]**	SLE and/or LN with 0 flares/year (N=162)	**Worse LIT scores** ($p=0.001$) and greater negative QoL impact with increased flare frequency
	SF-36 SCORES		
GLOBAL	**SLE + LN (N=566)[7]**	SLE only (N=1,261)	**Worse scores** in SF-36 BP and VT domains ($p\leq0.006$)
CHE	**Renal SLE (N=90)[8]**	Other SLE (N=162)	**Worse scores** in SF-36 RE ($p<0.001$), RP ($p=0.001$), BP ($p=0.02$), VT ($p=0.022$), SF domains ($p=0.007$)

Impact of Current SoC on HRQoL

- Only two studies were identified on the impact of currently available treatment options on patient HRQoL, both of which were published prior to 2015.
- No studies were identified on more recently approved therapies for LN, such as voclosporin or belimumab, due to age of identified studies.

Results (cont'd)

RITUXIMAB (RTX) ON HRQoL

- In a phase III RCT, RTX improved baseline SF-36 physical function score for patients with LN, although not statistically or nominally significant compared to mycophenolate mofetil (MMF) alone.[9]

CYCLOPHOSPHAMIDE (CYC) ON HRQoL

- One RWE study focused on outcomes of induction therapy with CYC and used lupus-specific QoL measures (SLE Symptom Checklist [SSC] and Influence of Rheumatic Diseases on General Health and Lifestyle [IRGL]) and the SF-36 questionnare.[10]
- Similar to results from previously published HRQoL studies in LN, fatigue was the most burdensome symptom at baseline (reported by >90% of patients with LN) and did not significantly improve after treatment.
- There was some observed improvement in certain domains of each lupus-specific PROM and an overall reduction in patient-reported distress and number of symptoms.[10]

Patient and Physician Satisfaction and HRQoL in Juvenile LN

PATIENT / PHYSICIAN PERSPECTIVES: Three studies examined the perspectives of patients with LN on satisfaction of disease control and treatment and facilitators of decision-making.[11-13]

- Despite currently available treatment options, dissatisfaction with disease control was reported by one fourth of nephrology physicians and one third of patients, with degree of dissatisfaction associated with LN severity and various signs and symptoms of disease.[11]
- LN patients noted that restoring health, improved QoL, and effective patient-physician communication regarding benefits and harms as the most relevant aspects of treatment.[12,13]

HRQoL IN JUVENILE POPULATION: From a single study on juvenile patients with LN, children on glucocorticoid therapy had reduced attention and worse problem internalization as reported by their parents using the Behavior Assessment of Childhood Disorders (BASC-2).[14]

Conclusions

- **Patients with LN suffer from poor HRQoL across multiple domains of life, especially during periods of active renal disease, compared to patients with SLE only.**
- **The limited evidence on HRQoL in patients with LN vs. patients with SLE only, measured by various lupus-specific PROMs in interventional studies, highlights a key evidence gap in understanding the humanistic burden of LN and the impact of SoC on patient well-being.**
- **1 in 3 patients with LN are dissatisfied with their disease control, further indicating a high unmet need for effective therapies and improved disease management.**

References

1. Yo JH, et al. Open Access Rheumatol. 2019;11:179-188. **2.** Kronbichler A, et al. Autoimmun Rev. 2019;18(5):510-518. **3.** Jolly M, et al. Lupus. 2018;27(2):257-264. **4.** Nieto R, et al. Arthritis Rheumatol. 2021; 73 (suppl 10). 5. Rogers JL, et al. Lupus Science & Medicine 2019;6. **6.**Katz P, et al. Qual Life Res. 2020;29(12):3251-3261. **7.** Hanly JG, et al. Rheumatology (Oxford). 2016;55(2):252-262. **8.** Chaigne B, et al. BMC Immunol. 2017;18(1):17. **9.** Rovin, BH, et al. Arthritis Rheum.2012;64(4):1215-26. **10.** Arends, S, et al. Netherlands Journal of Medicine.2014;72(9):481-490. 11. Mozaffarian N, et al. Patient Prefer Adherence. 2016;10:2051-2061. **12.** Singh JA, et al. Arthritis Res Ther. 2015;17:367. **13.** Qu H, et al. Arthritis Care Res (Hoboken). 2016;68(12):1787-1794. **14.** Knight A, et al. J Pediatr. 2017;189:181-188.e1.**15.** Stroie, AM, et al. Nephrology Dialysis Transplantation.2021;36(SUPPL 1):i188.

Author Disclosures and Acknowledgements

C. Arriens: GSK (honoraria), Aurinia (honoraria, speaker fees), BMS (honoraria, grant/research support), AstraZeneca (grant/research support, speaker fees); **J. Ma**: None; **K. Pisarczyk**: None; **R. Leff**: Kezar Life Sciences; **K. Palaniswamy**: Kezar Life Sciences; **L. Long**: Kezar Life Sciences.

Abbreviations: BASC-2, Behavior Assessment of Childhood Disorders; BP, bodily pain; CHE, Switzerland; CYC, cyclophosphamide; HRQoL, health-related quality of life; IRGL, Influence of Rheumatic Diseases on General Health and Lifestyle, LATAM, Latin America; LIT, Lupus Impact Tracker; LN, lupus nephritis; MMF, mycophenolate mofetil; PROM, patient-reported outcome measure; QoL, quality of life; RCT, randomized controlled trial; RE, role emotional; RP, role physical; RTX, rituximab; RWE, real-world evidence; SF, social functioning; SF-36, Short Form 36 Health Survey Questionnaire; SLAQ, Systemic Lupus Activity Questionnaire; SLE; systemic lupus erythematosus; SoC, standard of care; SSC, SLE Symptom Checklist; TI/AB, title and abstract; TLR, targeted literature review; USA, United States of America; VT, vitality.

Presented at American College of Rheumatology Convergence 2022, November 10-14, 2022. Abstract number: 1289849